Are Tryptase Thresholds For Systemic Mastocytosis Adequate? Electronic Health Record (EHR) Data Weigh In

Poster Number 527

Daniel S. Herman,[1] Mengmeng Li,[1] Philip Sarajlic,[1] Justin Tang,[1] Daniel Shaheen,[2] Pavle Milutinovic,[3] Elizabeth Hexner,[3] Olajumoke Fadugba[3]

[1]Department of Pathology & Laboratory Medicine, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [2]Blueprint Medicines Corporation, Cambridge, MA, USA; [3]Department of Medicine, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA

Introduction

- Systemic mastocytosis (SM) is an underdiagnosed clonal mast cell (MC) disease driven by the *KIT* D816V mutation in ~95% of cases,[1–4] and is characterized by the accumulation and abnormal activation of MCs which can lead to severe symptoms across multiple organ systems[5–7]
- A minor diagnostic criterion for SM per the World Health Organization classification is elevated serum tryptase (>20 ng/mL),[8] a serine protease enzyme that is produced by MCs,[9] which is an important mediator of inflammation and a marker of MC activation[10]
- At least 20% of patients with SM have only modestly elevated serum tryptase,[11] which limits the sensitivity of the SM diagnostic threshold (>20 ng/mL)[8]
- Hereditary α-tryptasemia (HαT) is a genetic trait defined by an increased copy number of the *TPSAB1* gene, which codes for α-tryptase, and can also lead to elevations in serum tryptase that overlap in magnitude with those seen with SM[12,13]
- HαT is the most common cause of elevated serum tryptase in the general population (~5%),[13] prompting calls to raise the "normal" tryptase range to improve specificity in identifying patients warranting further work-up for SM[14]
- Population studies have identified differences in the distribution of tryptase concentrations when grouping patients based on age or kidney function[15,16]
- Here, we have leveraged clinical data from a large, diverse US health system to evaluate whether conventional tryptase reference intervals were adequate

Methods

- We extracted EHR data for 6414 patients, independent of HαT status, in the Penn Medicine health system with initial ambulatory tryptase measurements from 2015 to 2024
- Reference intervals were indirectly estimated by fitting the predominant "normal" distribution of tryptase results to a Box-Cox-transformed Gaussian distribution (refineR v1.62).[17] This method enables robust estimation of reference intervals from routine clinical data
- *KIT* D816V in peripheral blood was measured by droplet digital PCR

Results

- Patients were median age 48 years (interquartile range: 34–63), 76% female, and 71% White. The majority of orders were placed by allergists and immunologists, followed by endocrinology, internal medicine, and hematologists/oncologists (**Table 1**)

Figure 1. Serum tryptase concentration distributions by specialty (A), eGFR (B), age (C), race (D), sex (E). Patients with testing ordered by hematologist/oncologists were excluded from (B-E) and those with kidney insufficiency (eGFR ≤60 mL/min/1.73 m²) were excluded from (C-E)

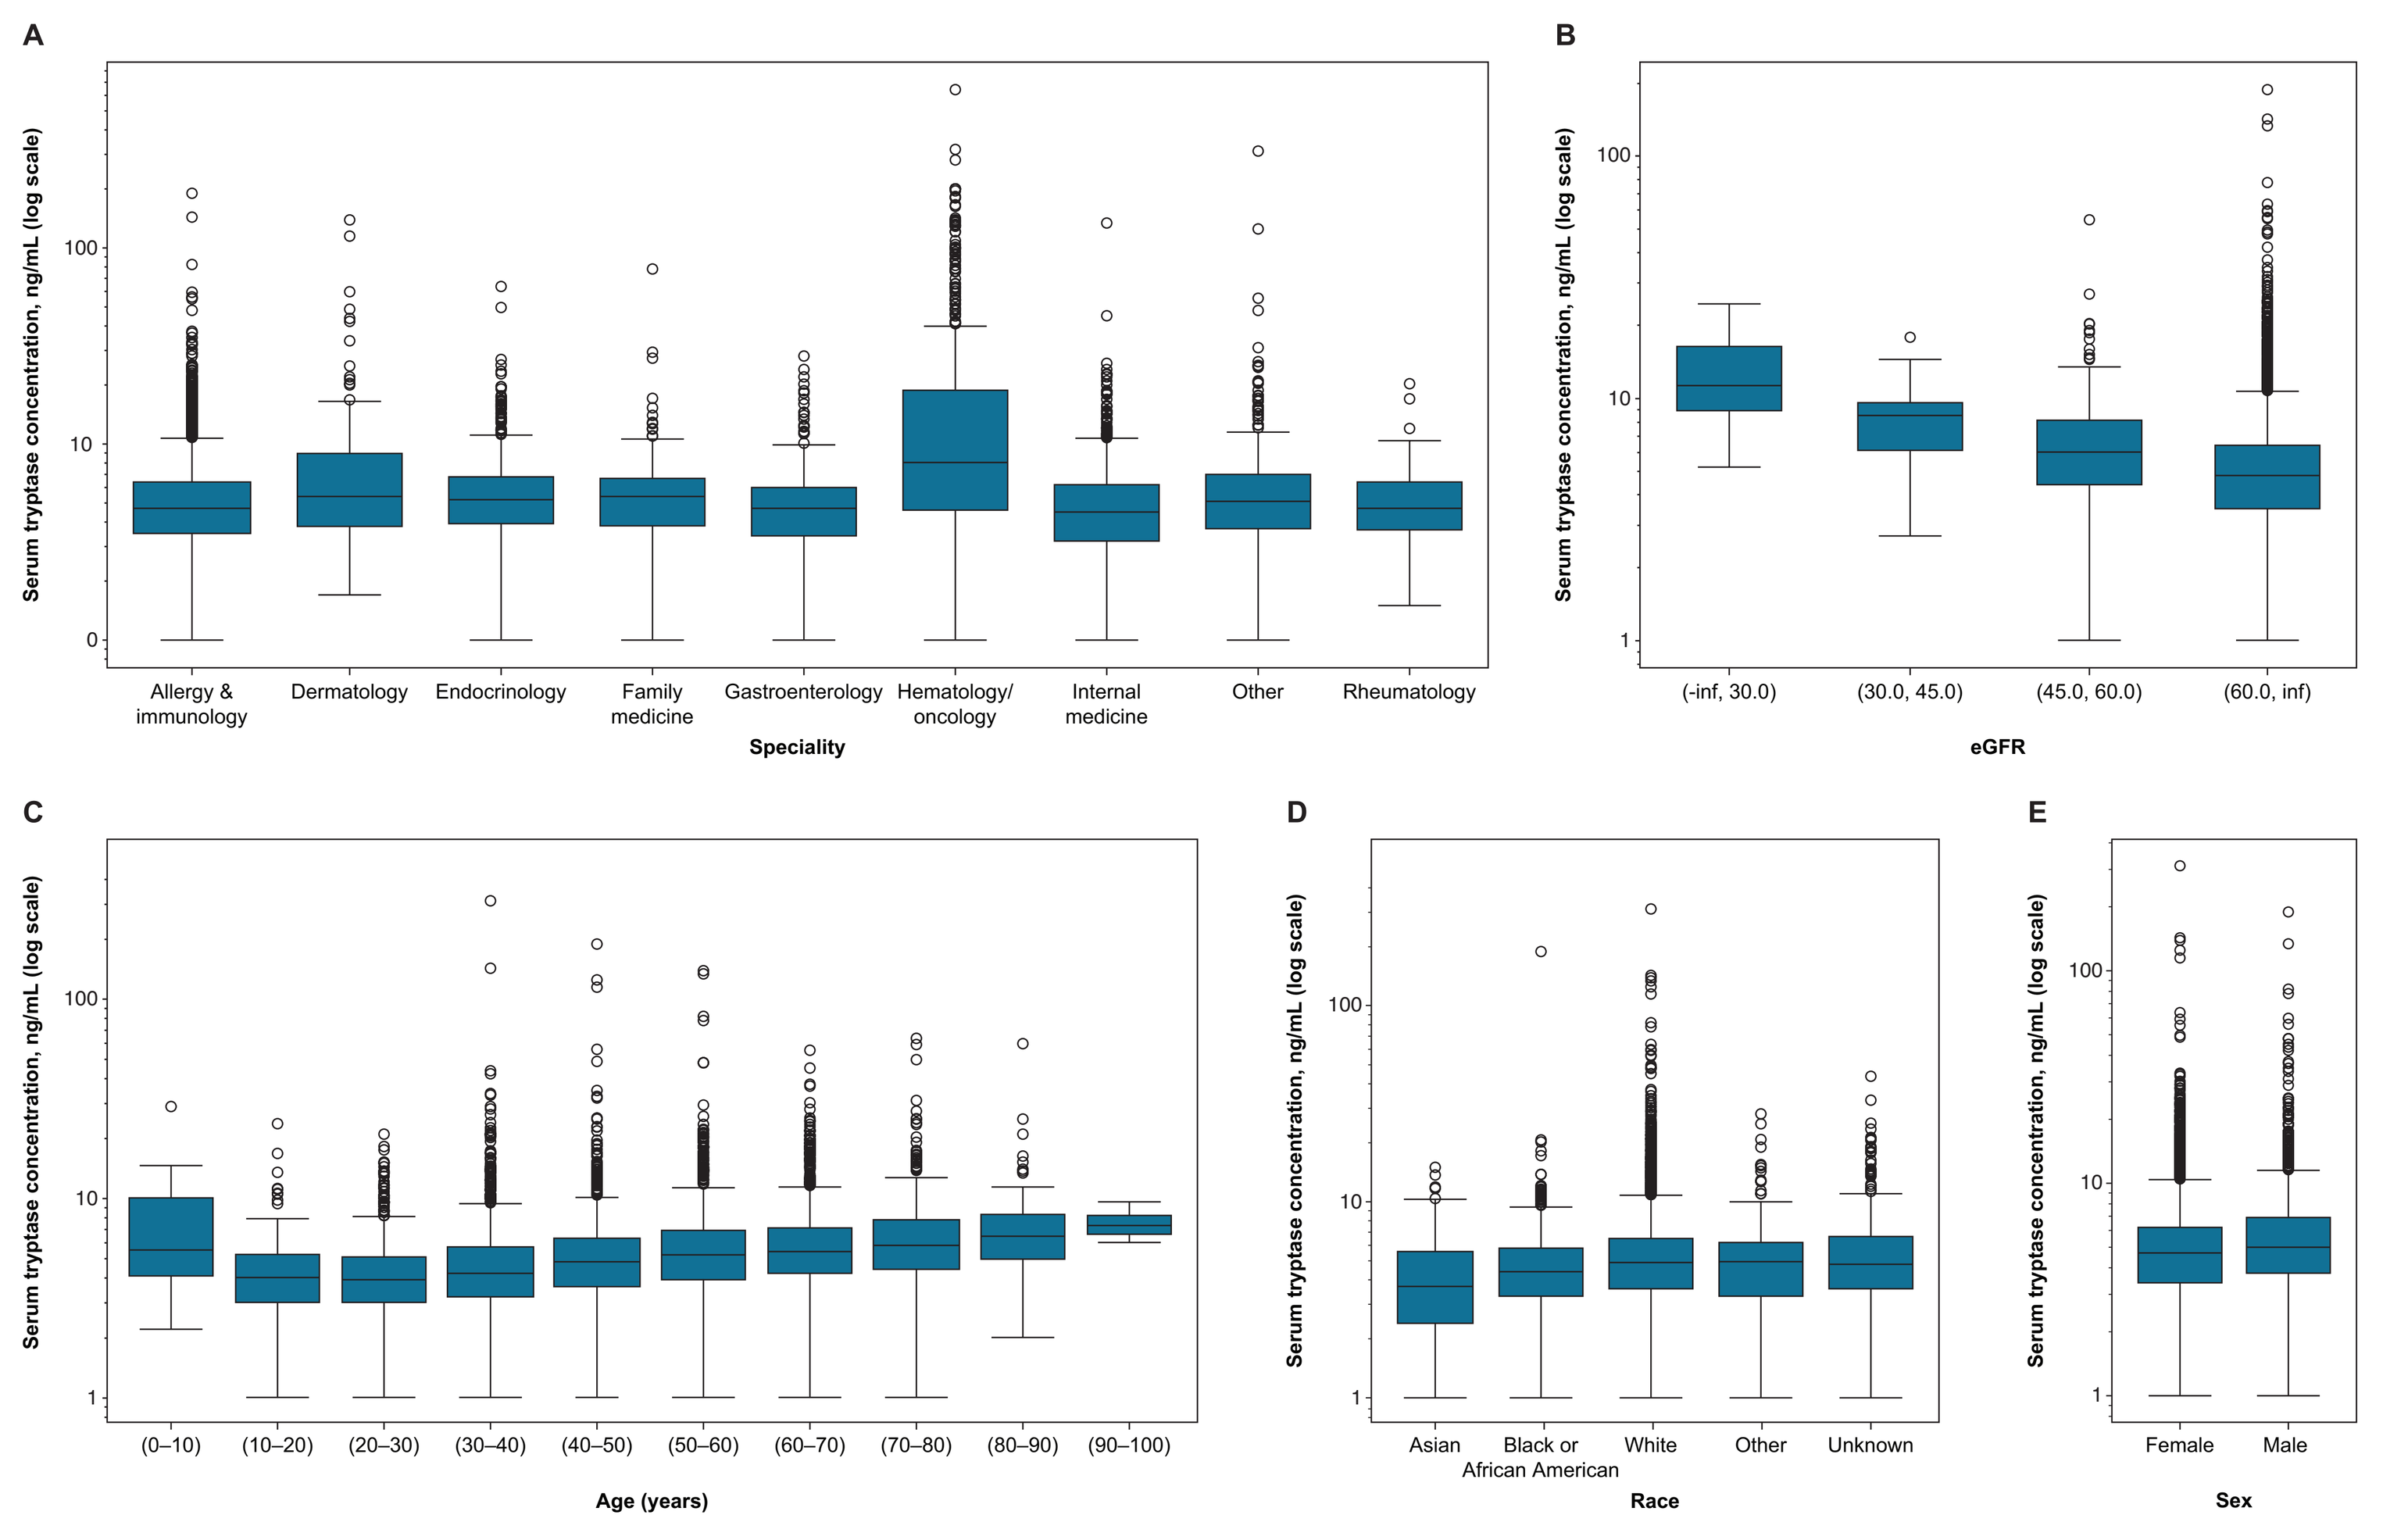

A
Serum tryptase concentration, ng/mL (log scale)
Allergy & immunology
Dermatology
Endocrinology
Family medicine
Gastroenterology
Hematology/oncology
Internal medicine
Other
Rheumatology
Speciality
B
(-inf, 30.0)
(30.0, 45.0)
(45.0, 60.0)
(60.0, inf)
eGFR
C
(0–10)
(10–20)
(20–30)
(30–40)
(40–50)
(50–60)
(60–70)
(70–80)
(80–90)
(90–100)
Age (years)
D
Asian
Black or African American
White
Other
Unknown
Race
E
Female
Male
Sex

- Serum tryptase concentrations were higher in patients whose testing was ordered by a hematology/oncology provider (**Figure 1A**), likely explained by multiple factors, including that patients with unrelated myeloid neoplasms have higher tryptase concentrations, there is an association between SM and some myeloid neoplasms, and patients referred to hematology/oncology may have more severe disease manifestations
- Tryptase distributions vary considerably by age and estimated glomerular filtration rate (eGFR), and slightly by race and sex (**Figure 1B-E**)

Table 1. Demographics and clinical characteristics

		Missing	Overall	Allergy & immunology	Dermatology	Endocrinology	Family medicine	Gastroenterology	Hematology/ oncology	Internal medicine	Other	Rheumatology
n			6414	3430	183	710	166	303	592	624	307	99
Age, median (Q1, Q3)		0	48.1 (33.7, 62.9)	44.4 (31.7, 59.1)	51.6 (37.9, 65.2)	63.8 (54.8, 69.6)	44.1 (31.9, 57.5)	42.9 (32.0, 57.9)	56.9 (40.4, 68.7)	46.8 (34.0, 61.3)	46.0 (32.0, 58.5)	43.4 (35.7, 54.6)
Sex, n (%)	**Female**		4846 (75.6)	2669 (77.8)	116 (63.4)	586 (82.5)	116 (69.9)	219 (72.3)	355 (60.0)	477 (76.4)	229 (74.6)	79 (79.8)
	Male		1568 (24.4)	761 (22.2)	67 (36.6)	124 (17.5)	50 (30.1)	84 (27.7)	237 (40.0)	147 (23.6)	78 (25.4)	20 (20.2)
Race, n (%)	**Asian**		331 (5.2)	207 (6.0)	8 (4.4)	31 (4.4)	4 (2.4)	7 (2.3)	22 (3.7)	33 (5.3)	15 (4.9)	4 (4.0)
	Black or African American		739 (11.5)	485 (14.1)	12 (6.6)	29 (4.1)	14 (8.4)	22 (7.3)	46 (7.8)	102 (16.3)	26 (8.5)	3 (3.0)
	White		4558 (71.1)	2238 (65.2)	142 (77.6)	603 (84.9)	126 (75.9)	260 (85.8)	460 (77.7)	422 (67.6)	226 (73.6)	81 (81.8)
	Other		229 (3.6)	129 (3.8)	7 (3.8)	18 (2.5)	10 (6.0)	4 (1.3)	14 (2.4)	31 (5.0)	12 (3.9)	4 (4.0)
	Unknown		495 (7.7)	329 (9.6)	12 (6.6)	25 (3.5)	10 (6.0)	7 (2.3)	46 (7.8)	33 (5.3)	28 (9.1)	5 (5.1)
	American Indian or Alaskan Native		13 (0.2)	11 (0.3)	0	1 (0.1)	0	0	0	1 (0.2)	0	0
	Patient Declined		35 (0.5)	21 (0.6)	2 (1.1)	3 (0.4)	2 (1.2)	2 (0.7)	3 (0.5)	1 (0.2)	0	1 (1.0)
	Native Hawaiian or Other Pacific Islander		13 (0.2)	9 (0.3)	0	0	0	1 (0.3)	1 (0.2)	1 (0.2)	0	1 (1.0)
	East Indian		1 (0)	1 (0)	0	0	0	0	0	0	0	0
Tryptase (ng/mL), median (Q1, Q3)		0	4.9 (3.6, 6.9)	4.7 (3.5, 6.4)	5.4 (3.8, 8.9)	5.2 (3.9, 6.8)	5.4 (3.8, 6.7)	4.7 (3.4, 6.0)	8.1 (4.6, 18.8)	4.5 (3.2, 6.2)	5.1 (3.7, 7.0)	4.7 (3.7, 6.4)
Blood *KIT* D816V, n (%)	**Detected**		45 (0.7)	5 (0.1)	4 (2.2)	1 (0.1)	0	0	31 (5.2)	0	4 (1.3)	0
	Missing		6143 (95.8)	3277 (95.5)	173 (94.5)	700 (98.6)	166 (100.0)	298 (98.3)	524 (88.5)	613 (98.2)	295 (96.1)	97 (98.0)
	Not Detected		226 (3.5)	148 (4.3)	6 (3.3)	9 (1.3)	0	5 (1.7)	37 (6.2)	11 (1.8)	8 (2.6)	2 (2.0)

Figure 2. Upper reference limit for serum tryptase estimated from all tested patients (A), excluding those with a first tryptase measurement ordered by a hematologist/oncologist provider (B), and further excluding patients with evidence of kidney insufficiency (eGFR ≤60 mL/min/1.73 m²) (C)

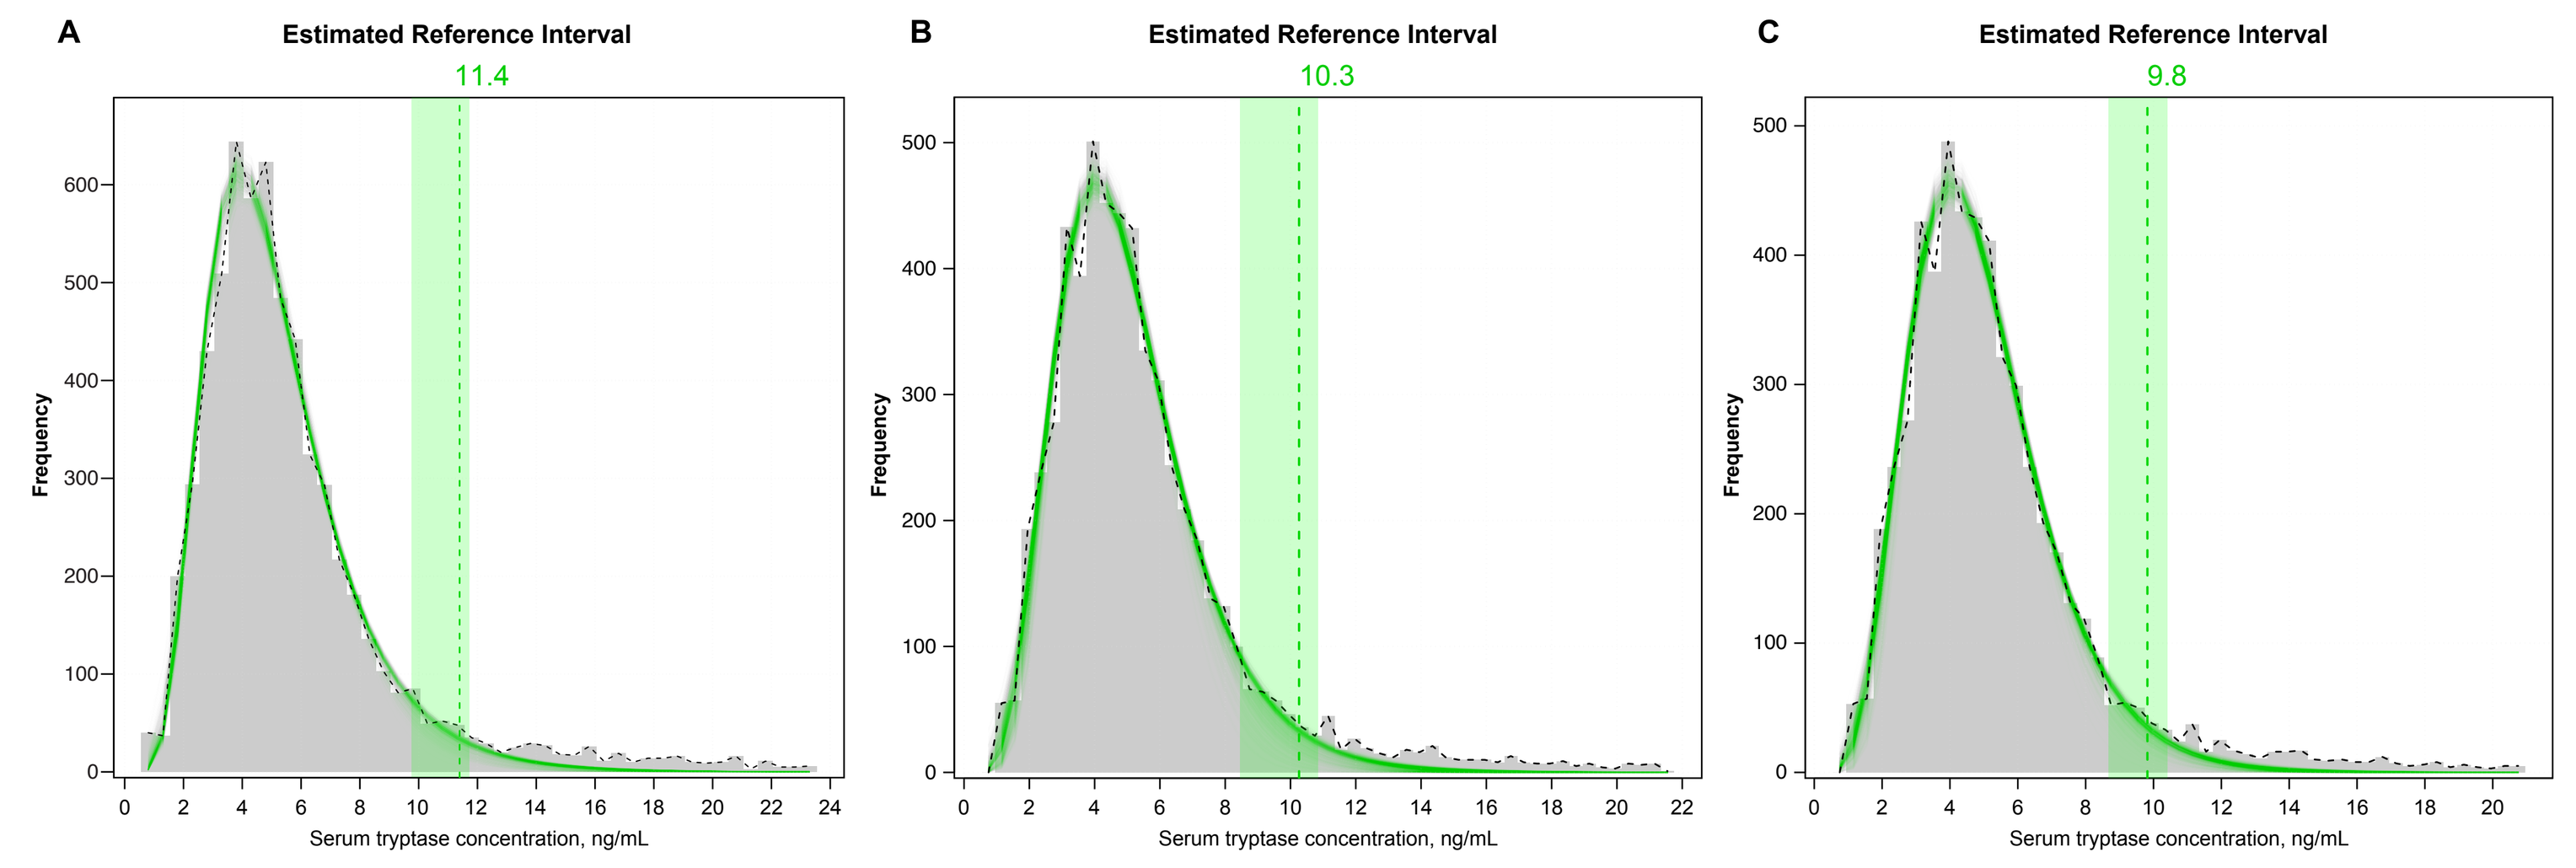

A
Estimated Reference Interval
11.4
B
Estimated Reference Interval
10.3
C
Estimated Reference Interval
9.8
Frequency
Serum tryptase concentration, ng/mL

The distribution of tryptase concentrations is shown as a gray histogram with a dashed black line. The green lines overlying the distribution represent the bootstrapped estimates for the "normal" distribution of tryptase. The estimated 97.5th percentile of these distributions is indicated with a vertical dashed green line and with green numeric label. The 95th percentile confidence interval for the estimated upper reference limit is shaded in green.

- Including all tested patients, the estimated serum tryptase reference upper limit (97.5th) was 11.4 ng/mL (95% confidence interval [CI]: 9.8–11.7; **Figure 2A**). To refine this estimate, we performed secondary analyses on subsets of the full cohort. We first excluded patients whose initial tryptase order was placed by a hematology/oncology provider, which yielded an estimated upper reference limit of 10.3 ng/mL (8.4–10.8; **Figure 2B**). Next, we further excluded patients with evidence of kidney insufficiency (eGFR ≤60 mL/min/1.73 m²), yielding an estimated upper reference limit of 9.8 ng/mL (8.7–10.4; **Figure 2C**)

Table 2. Detection of *KIT* D816V in peripheral blood

Serum tryptase (ng/mL)	No. of patients, n	No. w/*KIT* testing, n (%)	No. w/*KIT* detected, n (%)	Percent detected CI
≤8.4	5421	136 (2.5)	3 (2.2)	0.7–3.7%
8.5–9.8	257	15 (5.8)	1 (6.7)	0.0–13.3%
9.9–15.0	363	35 (9.6)	3 (8.6)	2.9–14.3%
>15.0	373	85 (22.8)	38 (44.7)	37.6–51.8%

- The proportions of patients with *KIT* D816V detected in peripheral blood increased with serum tryptase concentrations (**Table 2**). The patients with tryptase ≤8.4 ng/mL and *KIT* D816V mutation detected in peripheral blood had been tested because they had evidence of mastocytosis in the skin or bone marrow

Table 3. Likelihood of serum tryptase >9.8 ng/mL

Variable	Outcome	Test category	Ref category	Odds ratio	CI
Age	Tryptase >9.8	Age	per 10 years	1.30	1.23–1.38
Race	Tryptase >9.8	White	Non-White	1.86	1.47–2.42
Sex	Tryptase >9.8	Male	Female	1.54	1.25–1.92
eGFR	Tryptase >9.8	(45.0, 60.0)	(60.0, inf)	1.73	1.07–2.56
eGFR	Tryptase >9.8	(30.0, 45.0)	(60.0, inf)	3.46	1.54–6.72
eGFR	Tryptase >9.8	(-inf, 30.0)	(60.0, inf)	24.22	11.16–66.00

- Serum tryptase was >9.8 ng/mL more often with eGFR between 45 and 60 mL/min/1.73 m² (odds ratio [OR], 1.73 [1.07–2.56]), 30 and 45 (OR, 3.46 [1.54–6.72]), and <30 (OR, 24.22 [11.16–66.00]) (**Table 3**). Hematology/oncology patients were excluded from these analyses
- Excluding hematology/oncology patients and patients with eGFR ≤60 mL/min/1.73 m², tryptase was >9.8 ng/mL more often in male (OR, 1.54 [95% CI: 1.25–1.92]), White (OR, 1.86 [1.47–2.42]), and older patients (OR per 10 years, 1.30 [1.23–1.38]) (**Table 3**)
- We grouped the non-hematology/oncology, non-chronic kidney disease patients by age into 10–30 years old, 30–50 years old, 50–70 years old, 70–100 years old. Consistent with other reports, estimated indirect reference intervals (97.5th upper limit) appear to differ by age (**Figure 3**)

Figure 3. Estimated serum tryptase upper reference limits differ with age; 10–30 years old (A), 30–50 years old (B), 50–70 years old (C), and 70–100 years old (D)

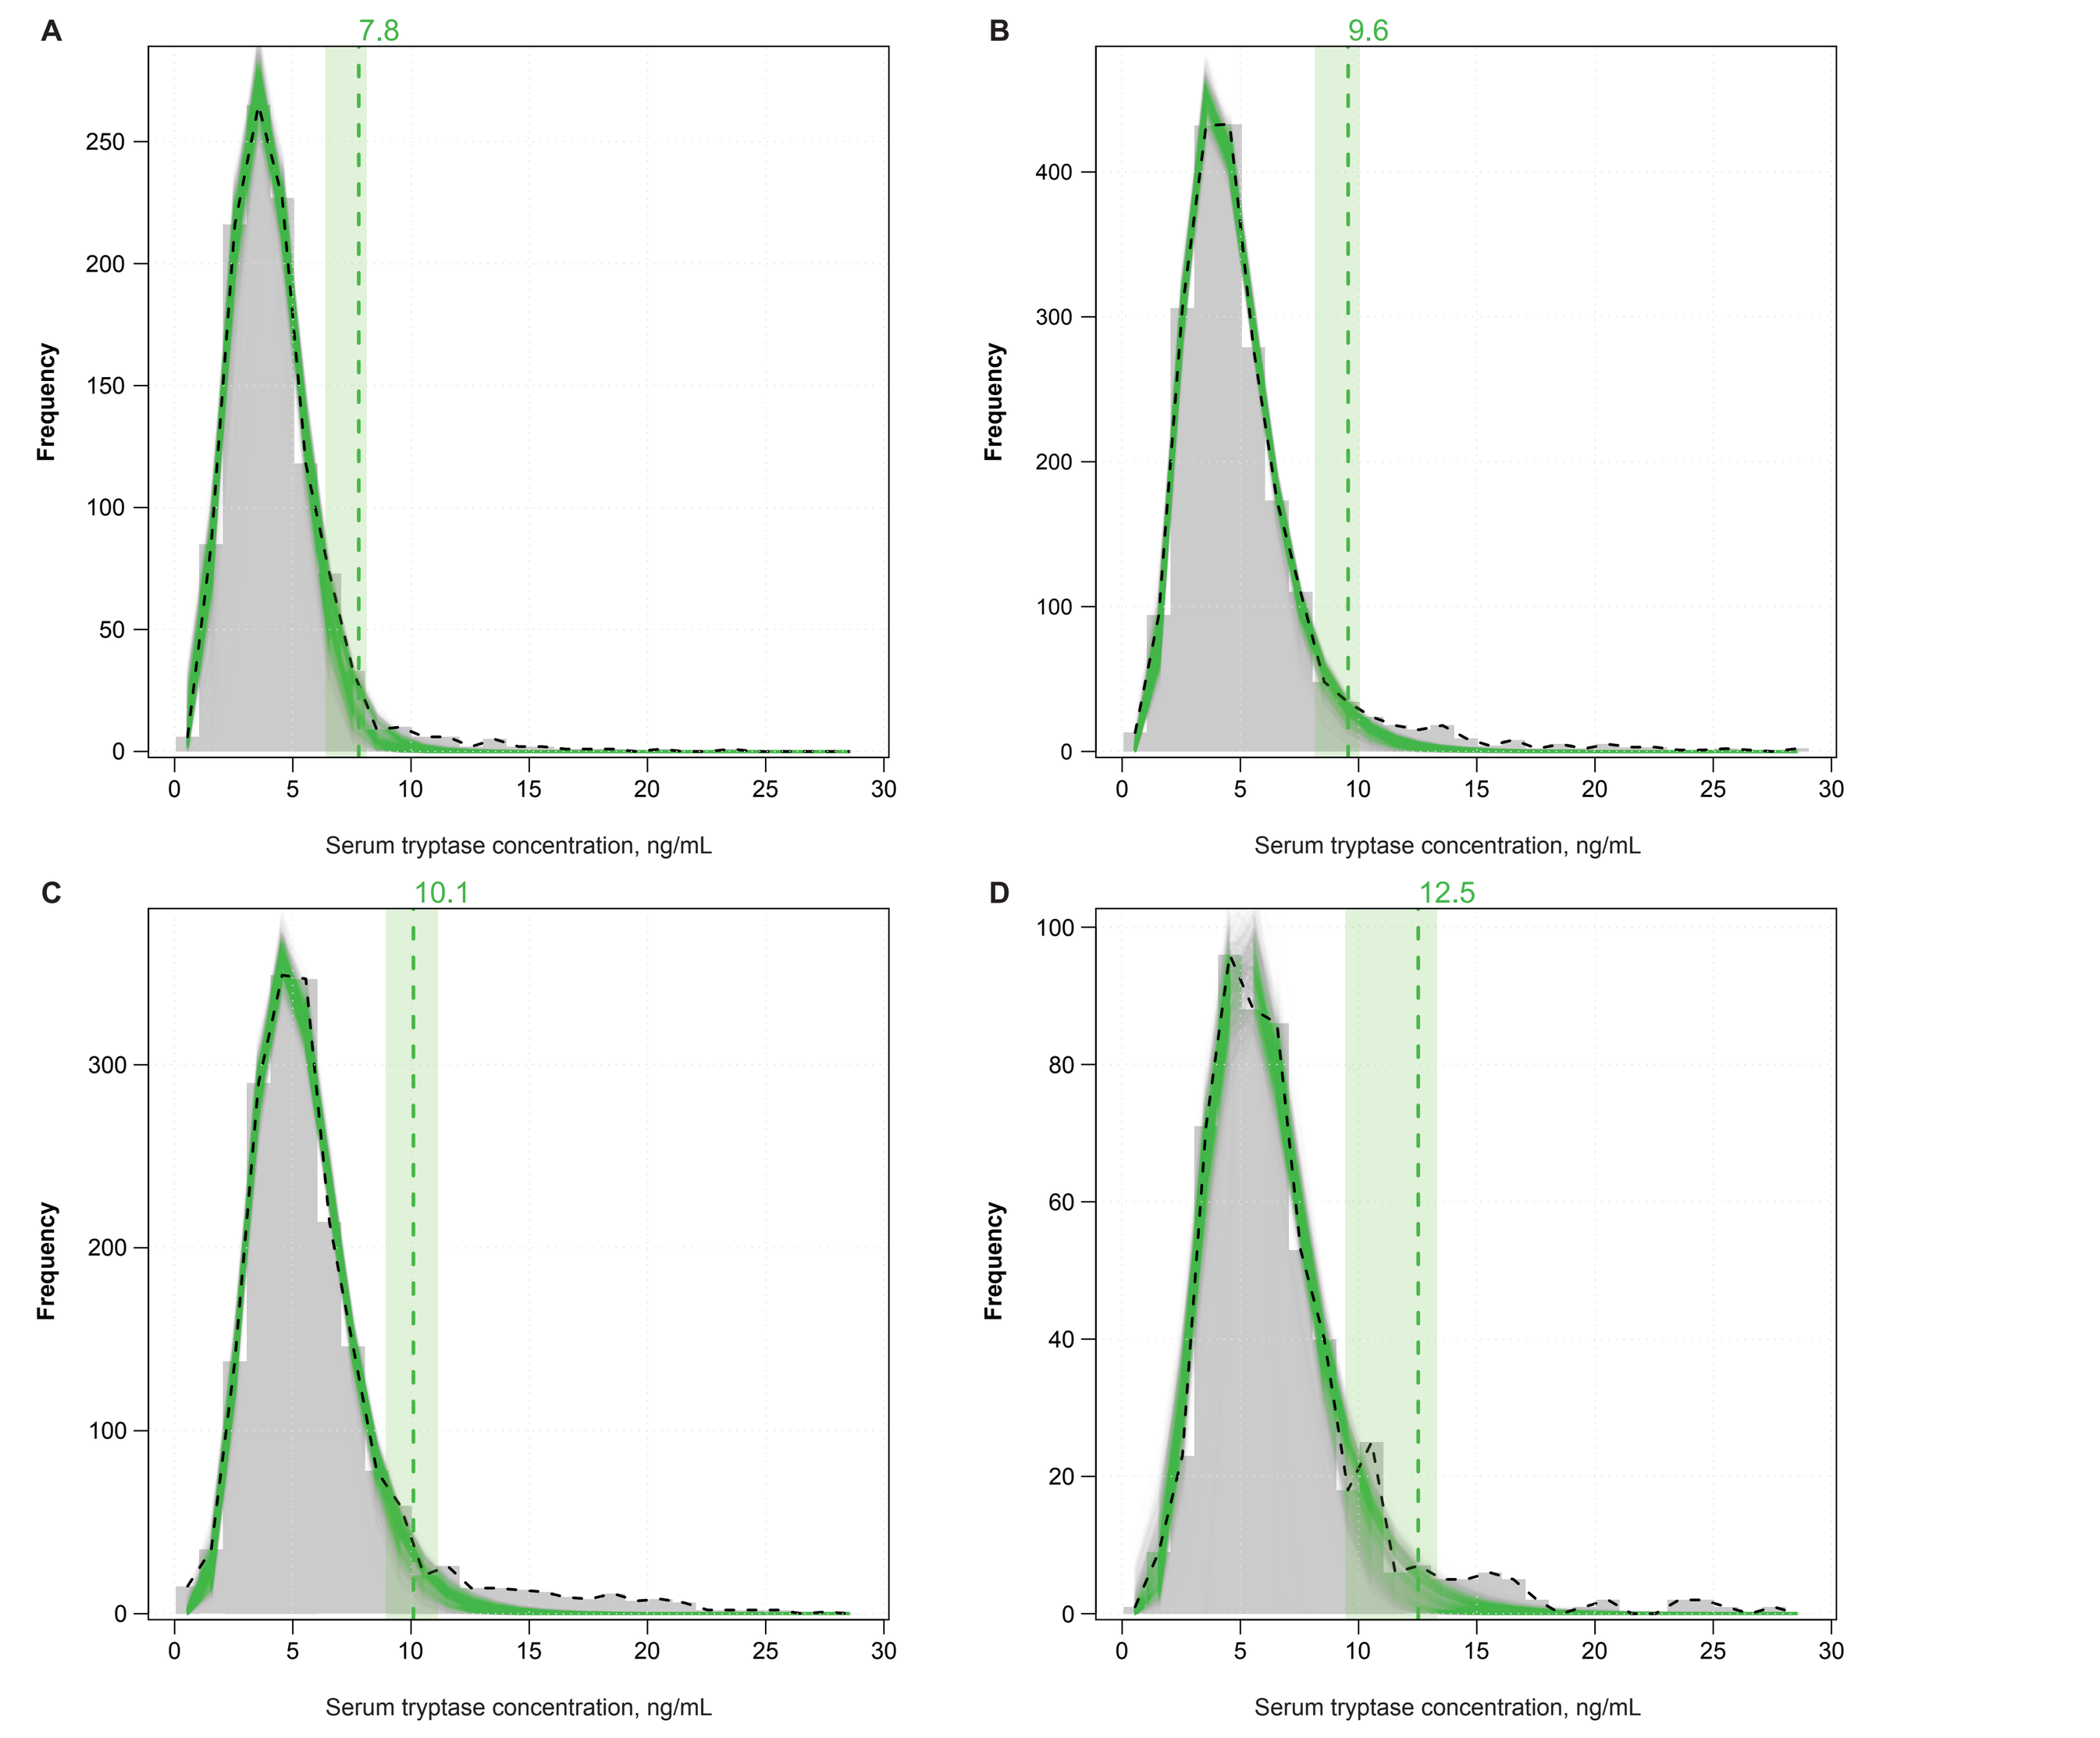

A
7.8
B
9.6
C
10.1
D
12.5
Frequency
Serum tryptase concentration, ng/mL

The distribution of tryptase concentrations is shown as a gray histogram with a dashed black line. The green lines overlying the distribution represent the bootstrapped estimates for the "normal" distribution of tryptase. The estimated 97.5th percentile of these distributions is indicated with a vertical dashed green line and with green numeric label. The 95th percentile confidence interval for the estimated upper reference limit is shaded in green.

Conclusions

- Retrospective EHR data analyses support maintaining conventional tryptase upper reference limits (~10 ng/mL) rather than raising them
- Age-specific reference intervals would likely improve the interpretation of tryptase results
- The derivation of tryptase reference intervals from EHR data should account for patients with myeloid neoplasms or kidney insufficiency
- Larger EHR studies should help refine interpretive thresholds
- *KIT* D816V results argue against using tryptase alone to rule out SM

Acknowledgements

Medical writing support was provided by Mark Lawrence, PhD, and editorial support was provided by Michelle Seddon, Dip Psychol, of the Prime Group of Companies (Knutsford, UK) and was funded by Blueprint Medicines Corporation, a wholly owned subsidiary of Sanofi. Responsibility for all opinions, conclusions, and data interpretation lies with the authors.

Scan the QR code to view our **Poster**

Conflicts of interest/disclosures

Daniel S. Herman has participated in investigator-initiated research sponsored by Blueprint Medicines Corporation, a wholly owned subsidiary of Sanofi.
For full author disclosures, please contact medinfo@blueprintmedicines.com

References

1. Kristensen T et al. *Am J Hematol*. 2014;89:493–498; 2. Garcia-Montero AC et al. *Blood*. 2006;108:2366–2372; 3. Ungerstedt J et al. *Cancers*. 2022;14:3942; 4. Pardanani A. *Am J Hematol*. 2023;98:1097–1116; 5. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 6. van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 7. Hartmann K et al. *J Allergy Clin Immunol*. 2016;137:35–45; 8. Khoury JD et al. *Leukemia*. 2022;36:1703–1719; 9. Wu R et al. *Curr Allergy Asthma Rep*. 2021;21:33; 10. Payne V et al. *Anaesthesia*. 2004;59:695–703; 11. McMurray JC et al. *Blood*. 2024;144:408–419; 12. González-de-Olano D et al. *Allergy*. 2024;79:711–723; 13. Lyons JJ et al. *J Allergy Clin Immunol*. 2021;147:622–632; 14. Valent P et al. *J Allergy Clin Immunol Pract*. 2023;11:3010–3020; 15. Chantran Y et al. *Allergy*. 2025;80:3217–3221; 16. Gonzalez-Quintela et al. *Clin Chem Lab Med*. 2010:48:701–706; 17. Ammer T et al. *Sci Rep*. 2021;11:16023.

Presented at the American Academy of Allergy, Asthma & Immunology (AAAAI) 2026 Annual Meeting, February 27–March 2, 2026, in Philadelphia, PA. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.